310

Outcomes in Older Patients With High-risk/Secondary AML Who Achieved Remission With CPX-351 Versus 7+3 but Did Not Undergo Transplant: Phase 3 Exploratory Analysis

American Society of Clinical Oncology (ASCO) Annual Meeting
May 29-31, 2020

Tara L. Lin,[1,*] Robert J. Ryan,[2] Stefan Faderl,[3] Ellen K. Ritchie[4]
[1]University of Kansas Medical Center, Kansas City, KS, USA; [2]Jazz Pharmaceuticals, Philadelphia, PA, USA; [3]Jazz Pharmaceuticals, Palo Alto, CA, USA; [4]Weill Cornell Medical College of Cornell University, New York, NY, USA.

*Presenting author.

Background

- Treatment outcomes with conventional induction chemotherapy (eg, 7+3 cytarabine/daunorubicin regimen) are particularly poor for older adults and those with high-risk/secondary AML[1-3]
- CPX-351 (Vyxeos®, daunorubicin and cytarabine liposome for injection), a dual-drug liposomal encapsulation of cytarabine/daunorubicin in a synergistic 5:1 molar ratio, has been approved by the US Food and Drug Administration and the European Medicines Agency for the treatment of adults with newly diagnosed, therapy-related AML (t-AML) or AML with myelodysplasia-related changes (AML-MRC)[4,5]
- Approval of CPX-351 was largely based on the primary endpoint analysis of a randomized phase 3 study that evaluated CPX-351 versus conventional 7+3 chemotherapy in older adults with newly diagnosed, high-risk/secondary AML
 - Induction followed by consolidation with CPX-351 significantly improved median overall survival (OS; primary endpoint) versus 7+3 (9.56 vs 5.95 months; hazard ratio [HR] = 0.69 [95% confidence interval (CI): 0.52, 0.90]; 1-sided $P = 0.003$)[6]
 - Of the patients who achieved remission, the CPX-351 arm had a higher rate of patients undergoing hematopoietic cell transplantation (HCT) versus the 7+3 arm (55% vs 46%), with median OS landmarked from the date of HCT not yet reached with CPX-351 versus 11.65 months with 7+3 (HR = 0.42 [95% CI: 0.20, 0.86])[7]
 - The safety profile of CPX-351 was consistent with the known safety profile of 7+3[6]
- The current analysis was conducted to better understand the impact of CPX-351 treatment on outcomes in patients who achieved remission but did not subsequently undergo HCT

Objective

- This exploratory *post hoc* analysis of the phase 3 study evaluated outcomes in the subgroup of patients who achieved complete remission (CR) or CR with incomplete neutrophil or platelet recovery (CRi) with CPX-351 versus 7+3 but did not undergo HCT

Methods

- Randomized, open-label, controlled, multicenter, phase 3 study (ClinicalTrials.gov Identifier: NCT01696084)[6]
- **Induction:** Patients were randomized 1:1 to receive 1 to 2 induction cycles with CPX-351 or 7+3, stratified by age (60 to 69 and 70 to 75 years) and AML subtype (see Key Inclusion Criteria)
 - CPX-351 arm: 100 units/m² (cytarabine 100 mg/m² + daunorubicin 44 mg/m²); 90-minute infusion on Days 1, 3, and 5 (Days 1 and 3 for second induction)
 - 7+3 arm: cytarabine 100 mg/m²/day continuous infusion for 7 days (5 days for second induction) + daunorubicin 60 mg/m² on Days 1, 2, and 3 (Days 1 and 2 for second induction)
- **Consolidation:** Up to 2 cycles of consolidation with CPX-351 65 units/m² (cytarabine 65 mg/m² + daunorubicin 29 mg/m²) or 5+2 chemotherapy for patients with CR or CRi
 - Patients could receive HCT at the discretion of the treating physician
- **Key Inclusion Criteria:** Adults 60 to 75 years of age; pathological diagnosis of AML according to 2008 World Health Organization criteria (≥20% blasts in peripheral blood or bone marrow); t-AML (based on prior cytotoxic treatment) or AML-MRC (history of myelodysplastic syndrome [MDS; with or without prior hypomethylating agents (HMAs)] or chronic myelomonocytic leukemia, or *de novo* AML with cytogenetic changes linked to MDS); ability to tolerate intensive AML chemotherapy in the opinion of the investigator; Eastern Cooperative Oncology Group (ECOG) performance status of 0 to 2
- **Key Exclusion Criteria:** Acute promyelocytic leukemia t(15;17) or favorable cytogenetics at screening; prior treatment intended as induction therapy for AML (hydroxyurea permitted); active secondary malignancies or central nervous system leukemia
- **Subgroup Analysis:** Patients enrolled in the phase 3 study who achieved CR or CRi during the study but did not undergo HCT were included in this subgroup analysis
 - The treatment groups were not prospectively balanced for this subgroup of patients

Results

- Of the 309 patients enrolled in the study, CR+CRi was achieved by 73/153 (48%) patients with CPX-351 versus 52/156 (33%) with 7+3
- Of the patients who achieved CR or CRi, 33/73 (45%) in the CPX-351 arm and 28/52 (54%) in the 7+3 arm did not subsequently undergo HCT

Table 1. Baseline Characteristics of Patients Who Achieved CR or CRi but Did Not Undergo HCT

Characteristic, n (%)	CPX-351 (n = 33)	7+3 (n = 28)
Demographic characteristics		
Age		
60 to 69 years, n (%)	20 (61)	17 (61)
70 to 75 years, n (%)	13 (39)	11 (39)
Male, n (%)	21 (64)	12 (43)
ECOG performance status, n (%)		
0	5 (15)	7 (25)
1	27 (82)	15 (54)
2	1 (3)	6 (21)
Clinical characteristics		
AML subtype, n (%)		
t-AML	6 (18)	5 (18)
AML with antecedent MDS		
With prior HMAs	7 (21)	11 (39)
Without prior HMAs	8 (24)	3 (11)
AML with antecedent CMML	2 (6)	3 (11)
de novo AML with MDS karyotype	10 (30)	6 (21)
Cytogenetic risk by NCCN, n (%)		
Favorable	4/31 (13)	5/25 (20)
Intermediate	13/31 (42)	10/25 (40)
Unfavorable	14/31 (45)	10/25 (40)
White blood cell count		
<20,000/µL	31 (94)	25 (89)
≥20,000/µL	2 (6)	3 (11)
Platelet count		
≤50,000/µL	24 (73)	15 (54)
>50,000/µL	9 (27)	13 (46)

CR, complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; HCT, hematopoietic cell transplantation; ECOG, Eastern Cooperative Oncology Group; AML, acute myeloid leukemia; t-AML, therapy-related acute myeloid leukemia; MDS, myelodysplastic syndrome; HMA, hypomethylating agent; CMML, chronic myelomonocytic leukemia; NCCN, National Comprehensive Cancer Network.

- The baseline characteristics of these patients were generally balanced between treatment arms; however, in the CPX-351 arm, there were more males and patients with an ECOG performance status of 1, but fewer patients with antecedent MDS and HMA exposure and with an ECOG performance status of 0 or 2 versus the 7+3 arm

Figure 1. OS in Patients Who Achieved CR or CRi but Did Not Undergo HCT

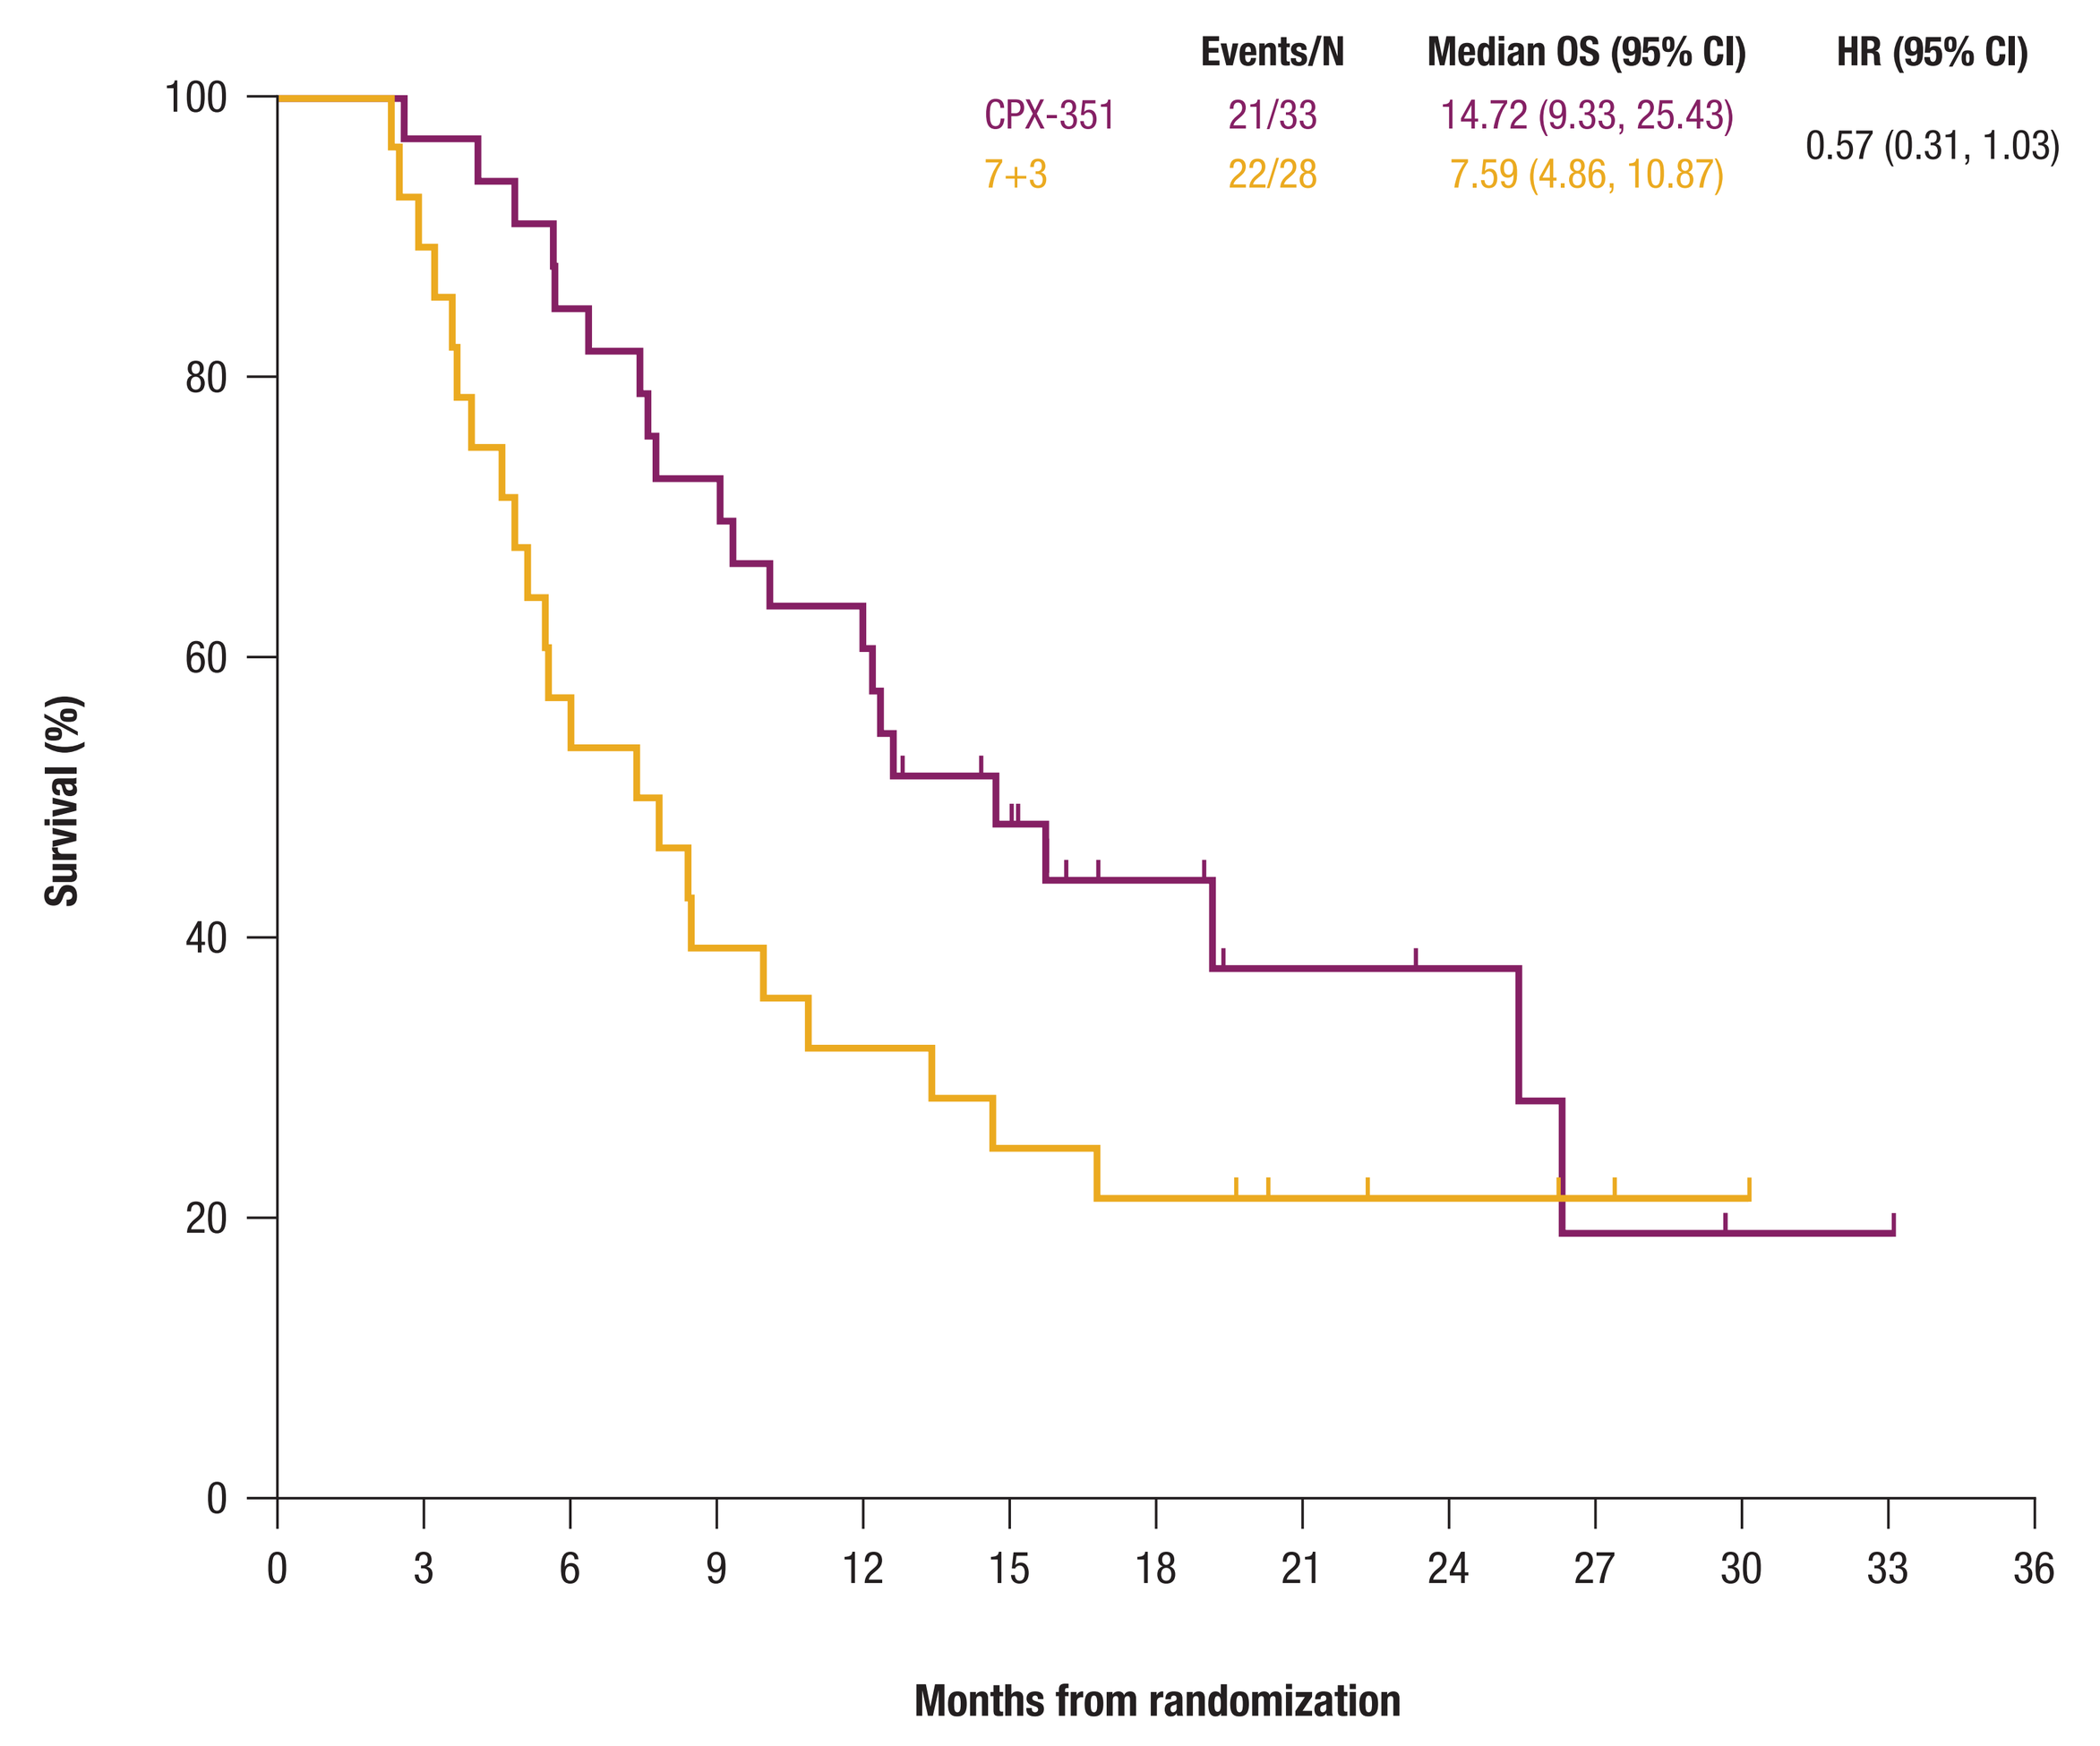
Events/N
Median OS (95% CI)
HR (95% CI)
CPX-351
21/33
14.72 (9.33, 25.43)
0.57 (0.31, 1.03)
7+3
22/28
7.59 (4.86, 10.87)
Survival (%)
100
80
60
40
20
0
0 3 6 9 12 15 18 21 24 27 30 33 36
Months from randomization

CPX-351	33	32	28	24	20	14	8	5	4	2	1	1	0
7+3	28	25	16	11	9	7	6	4	3	2	1	0	0

	CPX-351		7+3		
	n	Median OS, months	n	Median OS, months	HR (95% CI)
CR+CRi	33	14.72	28	7.59	0.57 (0.31, 1.03)
CR	27	19.15	21	8.41	0.55 (0.27, 1.10)
CRi	6	8.54	7	5.13	0.61 (0.18, 2.03)

OS, overall survival; CR, complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; HCT, hematopoietic cell transplantation; CI, confidence interval; HR, hazard ratio.

- Median OS was longer with CPX-351 versus 7+3 among patients who achieved CR or CRi but did not undergo HCT

Table 2. Summary of AEs in Patients Who Achieved CR or CRi but Did Not Undergo HCT

	CPX-351 (n = 33)	7+3 (n = 28)
Any AE, n (%)[a]	33 (100)	28 (100)
Febrile neutropenia	26 (79)	21 (75)
Nausea	21 (64)	13 (46)
Constipation	19 (58)	14 (50)
Diarrhea	17 (52)	20 (71)
Hypotension	16 (48)	6 (21)
Fatigue	15 (45)	10 (36)
Chills	14 (42)	9 (32)
Peripheral edema	14 (42)	14 (50)
Dyspnea	14 (42)	5 (18)
Cough	13 (39)	4 (14)
Rash	13 (39)	10 (36)
Abdominal pain	11 (33)	7 (25)
Mucosal inflammation	11 (33)	5 (18)
Dizziness	11 (33)	9 (32)
Insomnia	7 (21)	9 (32)
Pleural effusion	7 (21)	11 (39)
Any grade 3 or 4 AE, n (%)	31 (94)	20 (71)
Any serious AE, n (%)[b]	25 (76)	15 (54)
Febrile neutropenia	5 (15)	4 (14)
Acute respiratory failure	4 (12)	1 (4)
Sepsis	3 (9)	2 (7)
Ejection fraction decreased	3 (9)	1 (4)
Cardiac failure	2 (6)	1 (4)
Myocardial infarction	2 (6)	0
Syncope	2 (6)	0
Pneumonia	1 (3)	3 (11)
Atrial fibrillation	1 (3)	2 (7)
Subdural hemorrhage	0	2 (7)
Any grade 5 AE, n (%)	2 (6)	5 (18)
Discontinuation due to an AE, n (%)	1 (3)	0

AE, adverse event; CR, complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; HCT, hematopoietic cell transplantation.
[a]List of individual AEs includes all AEs occurring in ≥30% of patients in either treatment arm.
[b]List of individual AEs includes all serious AEs occurring in >1 patient in either treatment arm.

- The adverse event profile of CPX-351 among patients who achieved CR or CRi but did not undergo HCT was generally similar to that of conventional 7+3
- There was no early mortality by Day 60 in either treatment arm

Table 3. Median Time to Neutrophil and Platelet Recovery From Treatment Initiation in Patients Who Achieved CR or CRi but Did Not Undergo HCT

	CPX-351 (n = 33)	7+3 (n = 28)
Median (IQR) time to ANC recovery ≥500/µL, days	35 (29, 39)	29 (27.5, 35)
Median (IQR) time to platelet recovery ≥50,000/µL, days	36 (34, 42)	28.5 (27, 35)

CR, complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; HCT, hematopoietic cell transplantation; IQR, interquartile range; ANC, absolute neutrophil count.

- Median time to recovery of neutrophils and platelets was longer with CPX-351 than with 7+3 in patients who achieved CR or CRi with either 1 or 2 induction cycles

Conclusions

- CPX-351 improved median OS versus 7+3 in the subgroup of patients who achieved CR or CRi but did not undergo HCT, suggesting a benefit with CPX-351 treatment even among patients who did not subsequently undergo HCT
- The longer median OS in this subgroup of patients who achieved CR or CRi with CPX-351 versus conventional 7+3 suggests potentially deeper responses may be achieved with CPX-351 treatment
- The safety profile of CPX-351 in this subgroup was consistent with that of the overall study population[6] and the known safety profile of 7+3

References: 1. Granfeldt Østgård LS, et al. *J Clin Oncol.* 2015;33(31):3641-3649. **2.** Appelbaum FR, et al. *Blood.* 2006;107(9):3481-3485. **3.** Kayser S, et al. *Blood.* 2011;117(7):2137-2145. **4.** Vyxeos® (daunorubicin and cytarabine injection) liposome for injection, for intravenous use [package insert]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **5.** European Medicines Agency. Vyxeos liposomal 44 mg/100 mg powder for concentrate for solution for infusion. https://www.ema.europa.eu/en/medicines/human/EPAR/vyxeos-liposomal. Accessed April 8, 2020. **6.** Lancet JE, et al. *J Clin Oncol.* 2018;36(26):2684-2692. **7.** Faderl S, et al. Presented at: International Symposium on Acute Leukemias (ISAL) XVII; February 24-27, 2019; Munich, Germany. Poster 39.

Support: This study was supported by Jazz Pharmaceuticals.

Acknowledgments: Medical writing and editorial assistance were provided by Nancy Tang, PharmD, of SciFluent Communications, Inc., and were financially supported by Jazz Pharmaceuticals.

Disclosures: TLL has no relationships to disclose. **RJR** and **SF** are employees of and hold stock ownership/options in Jazz Pharmaceuticals. **EKR** has received consulting fees from Celgene, Incyte, Novartis, and Pfizer; has served on speakers bureaus for Ariad, Celgene, Incyte, and Novartis; has received research funding from Astellas Pharma, Bristol-Myers Squibb, Novartis, NS Pharma, and Pfizer; and has received travel support from Celgene and Novartis.

Contact information: For questions or comments, please contact Tara L. Lin at tlin@kumc.edu.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.