283

Five-year Final Results of a Phase 3 Study of CPX-351 Versus 7+3 in Older Adults With Newly Diagnosed High-risk/Secondary AML

American Society of Clinical Oncology (ASCO) Annual Meeting
May 29-31, 2020

Jeffrey E. Lancet,[1,*] Geoffrey L. Uy,[2] Laura F. Newell,[3] Tara L. Lin,[4] Ellen K. Ritchie,[5] Robert K. Stuart,[6] Stephen A. Strickland,[7] Donna Hogge,[8] Scott R. Solomon,[9] Dale L. Bixby,[10] Jonathan E. Kolitz,[11] Gary J. Schiller,[12] Matthew J. Wieduwilt,[13] Daniel H. Ryan,[14] Stefan Faderl,[15] Yu-Lin Chang,[15] Jorge E. Cortes[16,17]

[1]H. Lee Moffitt Cancer Center & Research Institute, Tampa, FL, USA; [2]Washington University School of Medicine, St. Louis, MO, USA; [3]Oregon Health & Science University, Portland, OR, USA; [4]University of Kansas Medical Center, Kansas City, KS, USA; [5]Weill Cornell Medical College of Cornell University, New York, NY, USA; [6]Hollings Cancer Center, Medical University of South Carolina, Charleston, SC, USA; [7]Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [8]Leukemia/BMT Program of British Columbia, Vancouver, BC, Canada; [9]Leukemia Program, Northside Hospital Cancer Center Institute, Atlanta, GA, USA; [10]Comprehensive Cancer Center, University of Michigan, Grass Lake, MI, USA; [11]Monter Cancer Center, Northwell Health System, Lake Success, NY, USA; [12]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [13]University of California – San Diego Moores Cancer Center, La Jolla, CA, USA; [14]University of Rochester, Rochester, NY, USA; [15]Jazz Pharmaceuticals, Palo Alto, CA, USA; [16]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [17]Georgia Cancer Center, Augusta University, Augusta, GA, USA.

*Presenting author.

Background

- Outcomes for the treatment of acute myeloid leukemia (AML) with conventional induction chemotherapy (eg, 7+3 cytarabine/daunorubicin regimen) are particularly poor for older adults and those with high-risk/secondary AML[1-3]
- CPX-351 (Vyxeos®; daunorubicin and cytarabine liposome for injection), a dual-drug liposomal encapsulation of cytarabine/daunorubicin in a synergistic 5:1 molar ratio, has been approved by the US Food and Drug Administration and the European Medicines Agency for the treatment of adults with newly diagnosed, therapy-related AML (t-AML) or AML with myelodysplasia-related changes (AML-MRC)[4,5]
- The primary endpoint analysis of the pivotal phase 3 trial (ClinicalTrials.gov Identifier: NCT01696084) that formed the basis for these approvals evaluated patients aged 60 to 75 years with newly diagnosed high-risk/secondary AML[6]
 - After a median follow-up of 20.7 months, CPX-351 significantly improved median overall survival (OS; primary endpoint) versus conventional 7+3 (9.56 vs 5.95 months; hazard ratio = 0.69 [95% confidence interval: 0.52, 0.90]; 1-sided $P = 0.003$)[6]
 - The overall safety profile of CPX-351 was consistent with the known safety profile of conventional 7+3 chemotherapy[6]

Objective

- The objective of the current analysis was to report the prospectively planned, final 5-year follow-up results from this phase 3 trial

Methods

- Randomized, open-label, controlled, multicenter, phase 3 study (ClinicalTrials.gov Identifier: NCT01696084)[6]
- **Induction:** Patients were randomized 1:1 to receive 1 to 2 induction cycles with CPX-351 or 7+3, stratified by age (60 to 69 and 70 to 75 years) and AML subtype (see Inclusion Criteria)
 - CPX-351 arm: 100 units/m^2 (cytarabine 100 mg/m^2 + daunorubicin 44 mg/m^2); 90-minute infusion on Days 1, 3, and 5 (Days 1 and 3 for second induction)
 - 7+3 arm: cytarabine 100 mg/m^2/day continuous infusion for 7 days (5 days for second induction) + daunorubicin 60 mg/m^2 on Days 1, 2, and 3 (Days 1 and 2 for second induction)
- **Consolidation:** Up to 2 cycles of consolidation with CPX-351 65 units/m^2 (cytarabine 65 mg/m^2 + daunorubicin 29 mg/m^2) or 5+2 chemotherapy for patients with complete remission (CR) or CR with incomplete neutrophil or platelet recovery (CRi)
 - Patients could receive hematopoietic cell transplantation (HCT) at the discretion of the treating physician
- **Key Inclusion Criteria:** Adults 60 to 75 years of age; pathological diagnosis of AML according to 2008 World Health Organization criteria (≥20% blasts in peripheral blood or bone marrow); t-AML (based on prior cytotoxic treatment) or AML-MRC (history of myelodysplastic syndrome [MDS; with or without prior hypomethylating agents] or chronic myelomonocytic leukemia, or *de novo* AML with cytogenetic changes linked to MDS); ability to tolerate intensive AML chemotherapy in the opinion of the investigator; Eastern Cooperative Oncology Group (ECOG) performance status of 0 to 2
- **Key Exclusion Criteria:** Acute promyelocytic leukemia t(15;17) or favorable cytogenetics at screening; prior treatment intended as induction therapy for AML (hydroxyurea permitted); active secondary malignancies or central nervous system leukemia
- Patients were followed until death or up to 5 years following randomization

Results

- In total, 309 patients were randomized to receive CPX-351 (n = 153) or 7+3 (n = 156) and were included in the intent-to-treat population for efficacy analyses; the safety population included 153 and 151 patients, respectively[6]

Table 1. Baseline Characteristics in Older Adults With Newly Diagnosed High-risk/Secondary AML[6]

Characteristic, n (%)	CPX-351 (n = 153)	7+3 (n = 156)
Demographic characteristics		
Age		
Mean (SD), years	67.8 (4.2)	67.7 (4.1)
60 to 69 years, n (%)	96 (63)	102 (65)
70 to 75 years, n (%)	57 (37)	54 (35)
Male, n (%)	94 (61)	96 (62)
ECOG performance status, n (%)		
0	37 (24)	45 (29)
1	101 (66)	89 (57)
2	15 (10)	22 (14)
Clinical characteristics		
AML subtype, n (%)		
t-AML	30 (20)	33 (21)
AML with antecedent MDS		
With prior HMAs	50 (33)	55 (35)
Without prior HMAs	21 (14)	19 (12)
AML with antecedent CMML	11 (7)	12 (8)
de novo AML with MDS karyotype	41 (27)	37 (24)
Prior HMA therapy, n (%)[a]	62 (41)	71 (46)
Cytogenetic risk by NCCN, n (%)	143	146
Favorable	7 (5)	5 (3)
Intermediate	64 (45)	58 (40)
Unfavorable	72 (50)	83 (57)
Median bone marrow blasts (range), %	35 (5, 93)	35 (3, 97)
WBC count <20,000/μL, n (%)	131/153 (86)	131/155 (85)

AML, acute myeloid leukemia; SD, standard deviation; ECOG, Eastern Cooperative Oncology Group; t-AML, therapy-related AML; MDS, myelodysplastic syndrome; HMA, hypomethylating agent; CMML, chronic myelomonocytic leukemia; NCCN, National Comprehensive Cancer Network; WBC, white blood cell.
[a]Includes patients in the prespecified randomization strata of antecedent MDS with prior HMA exposure as well as patients in other strata (eg, t-AML, antecedent CMML) who had previously received HMAs.

Table 2. Multivariable Cox Regression Analysis of Baseline Factors Associated With OS

	n/N (%)	HR (95% CI)
ECOG performance status, n (%)		
0	72/287 (25)	0.53 (0.33, 0.84)
1	180/287 (63)	0.72 (0.48, 1.08)
2	35/287 (12)	
Karyotype		
Non-poor	133/287 (46)	0.49 (0.36, 0.66)
Poor	154/287 (54)	
WBC count		
<20 × 10^9/L	174/287 (61)	1.64 (1.25, 2.16)
≥20 × 10^9/L	113/287 (39)	
Platelet count		
≤50 × 10^9/L	246/287 (86)	0.64 (0.44, 0.93)
>50 × 10^9/L	41/287 (14)	
Treatment arm		
CPX-351	143/287 (50)	0.70 (0.53, 0.92)
7+3	144/287 (50)	

OS, overall survival; HR, hazard ratio; CI, confidence interval; ECOG, Eastern Cooperative Oncology Group; WBC, white blood cell.

- A univariable analysis identified ECOG performance status, cytogenetic risk, white blood cell count, platelet count, and treatment arm as having a significant association with the primary endpoint, OS
 - Covariates not found to be associated with OS included sex, hemoglobin levels, percent bone marrow blasts, and presence of an *FLT3*-ITD mutation
- Factors that were found to be associated with longer OS in a multivariable analysis included lower ECOG performance status, non-poor karyotype, lower white blood cell count, higher platelet count, and treatment with CPX-351

Figure 1. OS in Older Adults With Newly Diagnosed High-risk/Secondary AML

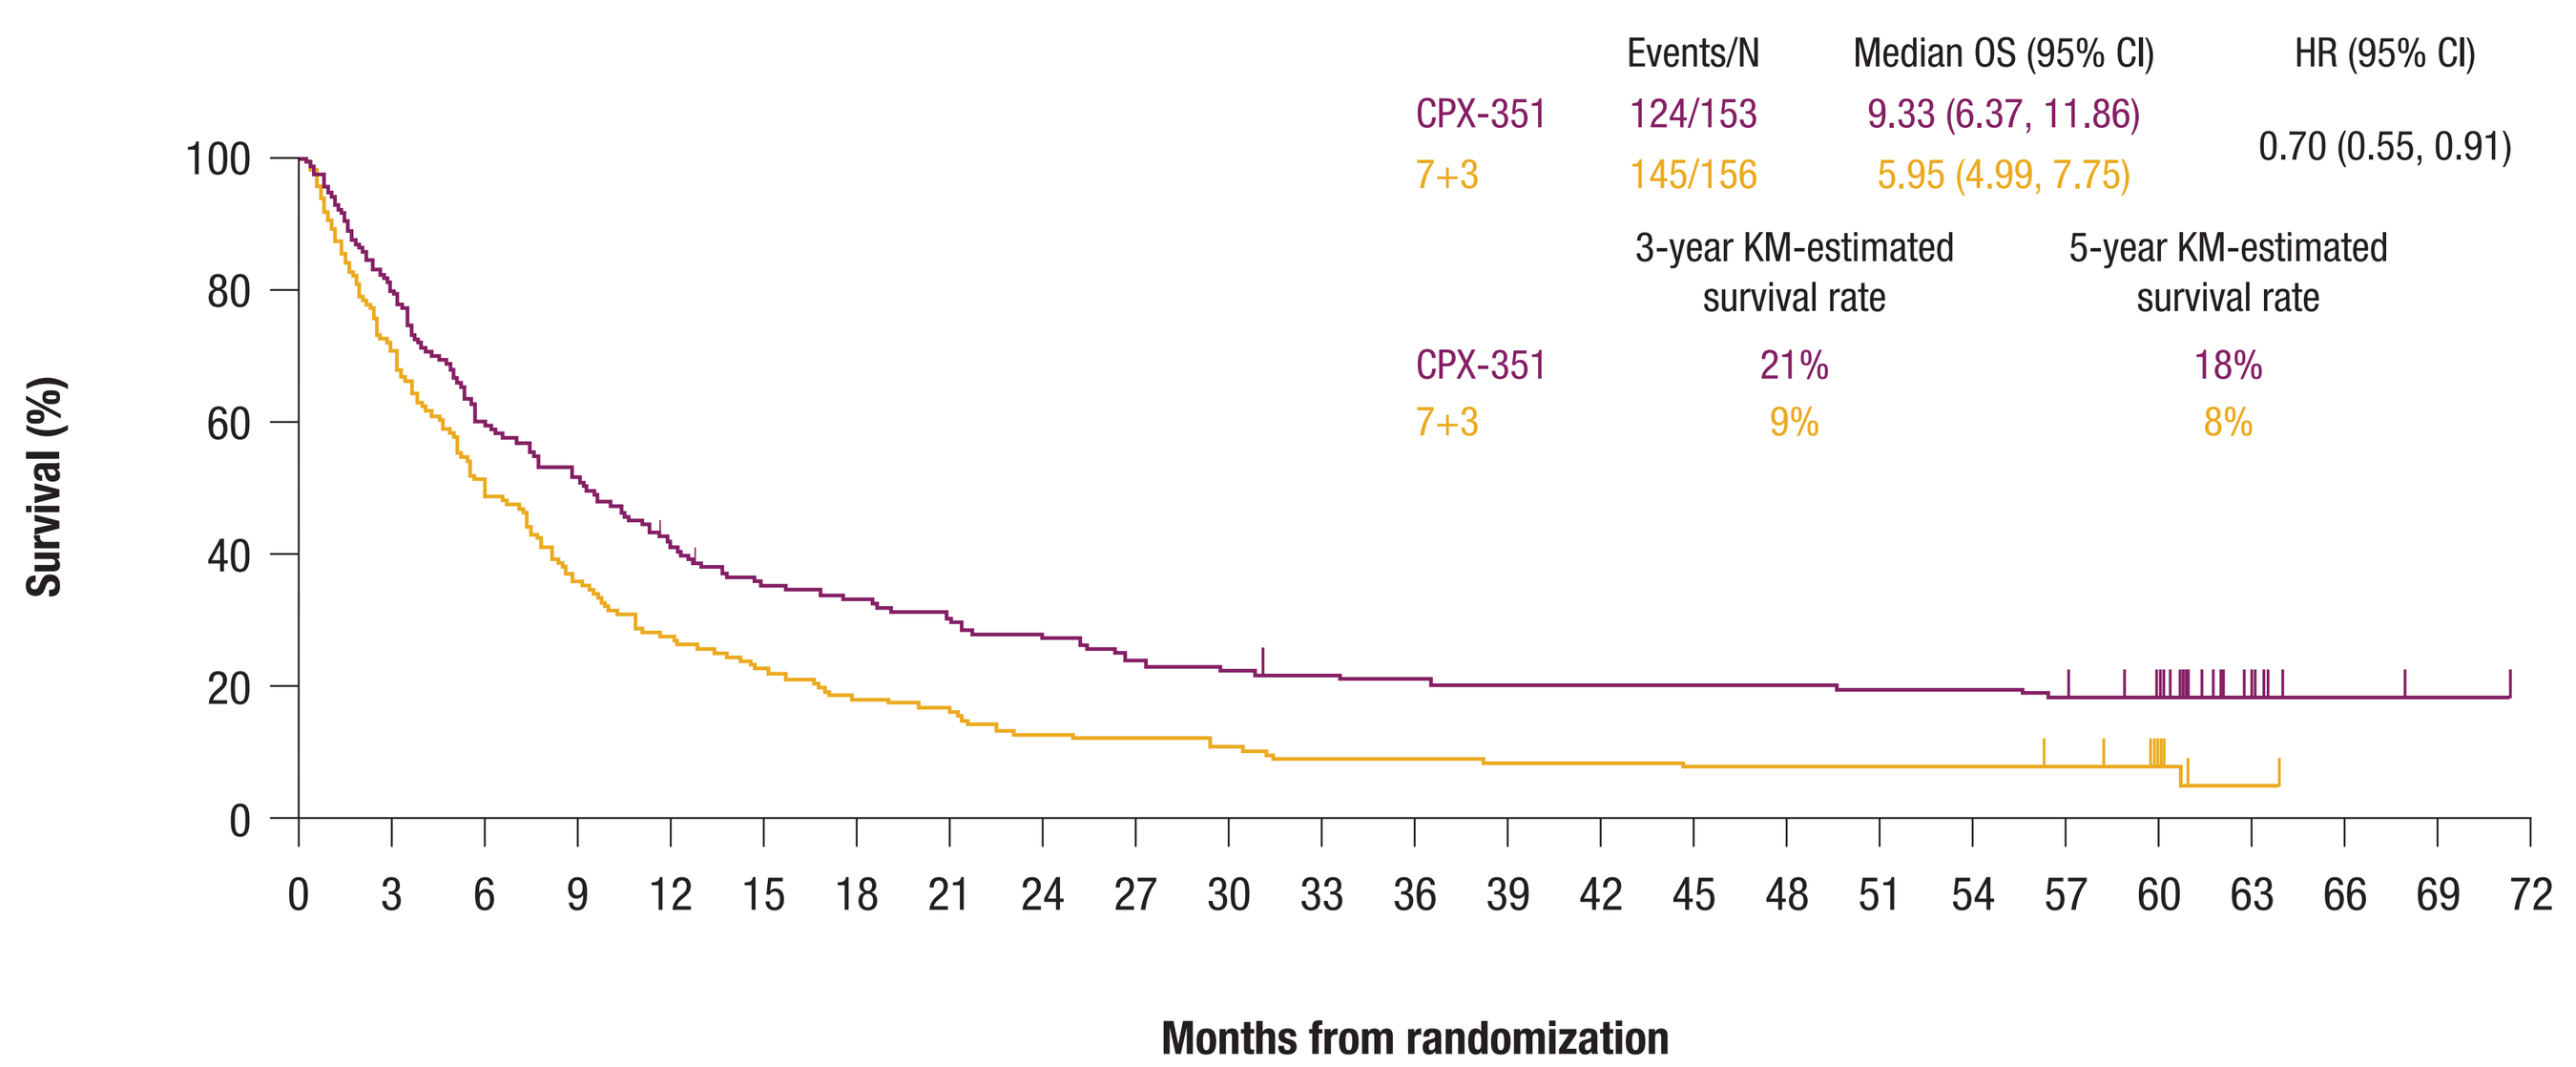

Events/N
Median OS (95% CI)
HR (95% CI)
CPX-351
124/153
9.33 (6.37, 11.86)
7+3
145/156
5.95 (4.99, 7.75)
0.70 (0.55, 0.91)
3-year KM-estimated survival rate
5-year KM-estimated survival rate
CPX-351
21%
18%
7+3
9%
8%
Survival (%)
100
80
60
40
20
0
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51 54 57 60 63 66 69 72
Months from randomization

CPX-351	153	122	92	79	62	52	49	45	40	35	33	31	30	29	29	29	29	28	28	26	22	6	2	1	0
7+3	156	110	77	56	43	35	28	25	20	19	17	14	14	13	13	12	12	12	12	11	5	1	0	0	0

OS, overall survival; AML, acute myeloid leukemia; CI, confidence interval; HR, hazard ratio; KM, Kaplan-Meier.

- After a median follow-up of 60.65 months (10th to 90th percentile: 58.22, 63.90), improved median OS with CPX-351 versus 7+3 was maintained, with a hazard ratio that was very stable and consistent with the prior primary endpoint analysis
 - The Kaplan-Meier–estimated survival rates were higher for CPX-351 versus 7+3 at 3 years (21% vs 9%) and 5 years (18% vs 8%)
 - Median OS for the CPX-351 arm differed from that reported for the primary endpoint analysis due to a patient death reported after the cut-off date for that analysis

Figure 2. OS Landmarked From the Date of HCT in Older Adults With Newly Diagnosed High-risk/Secondary AML

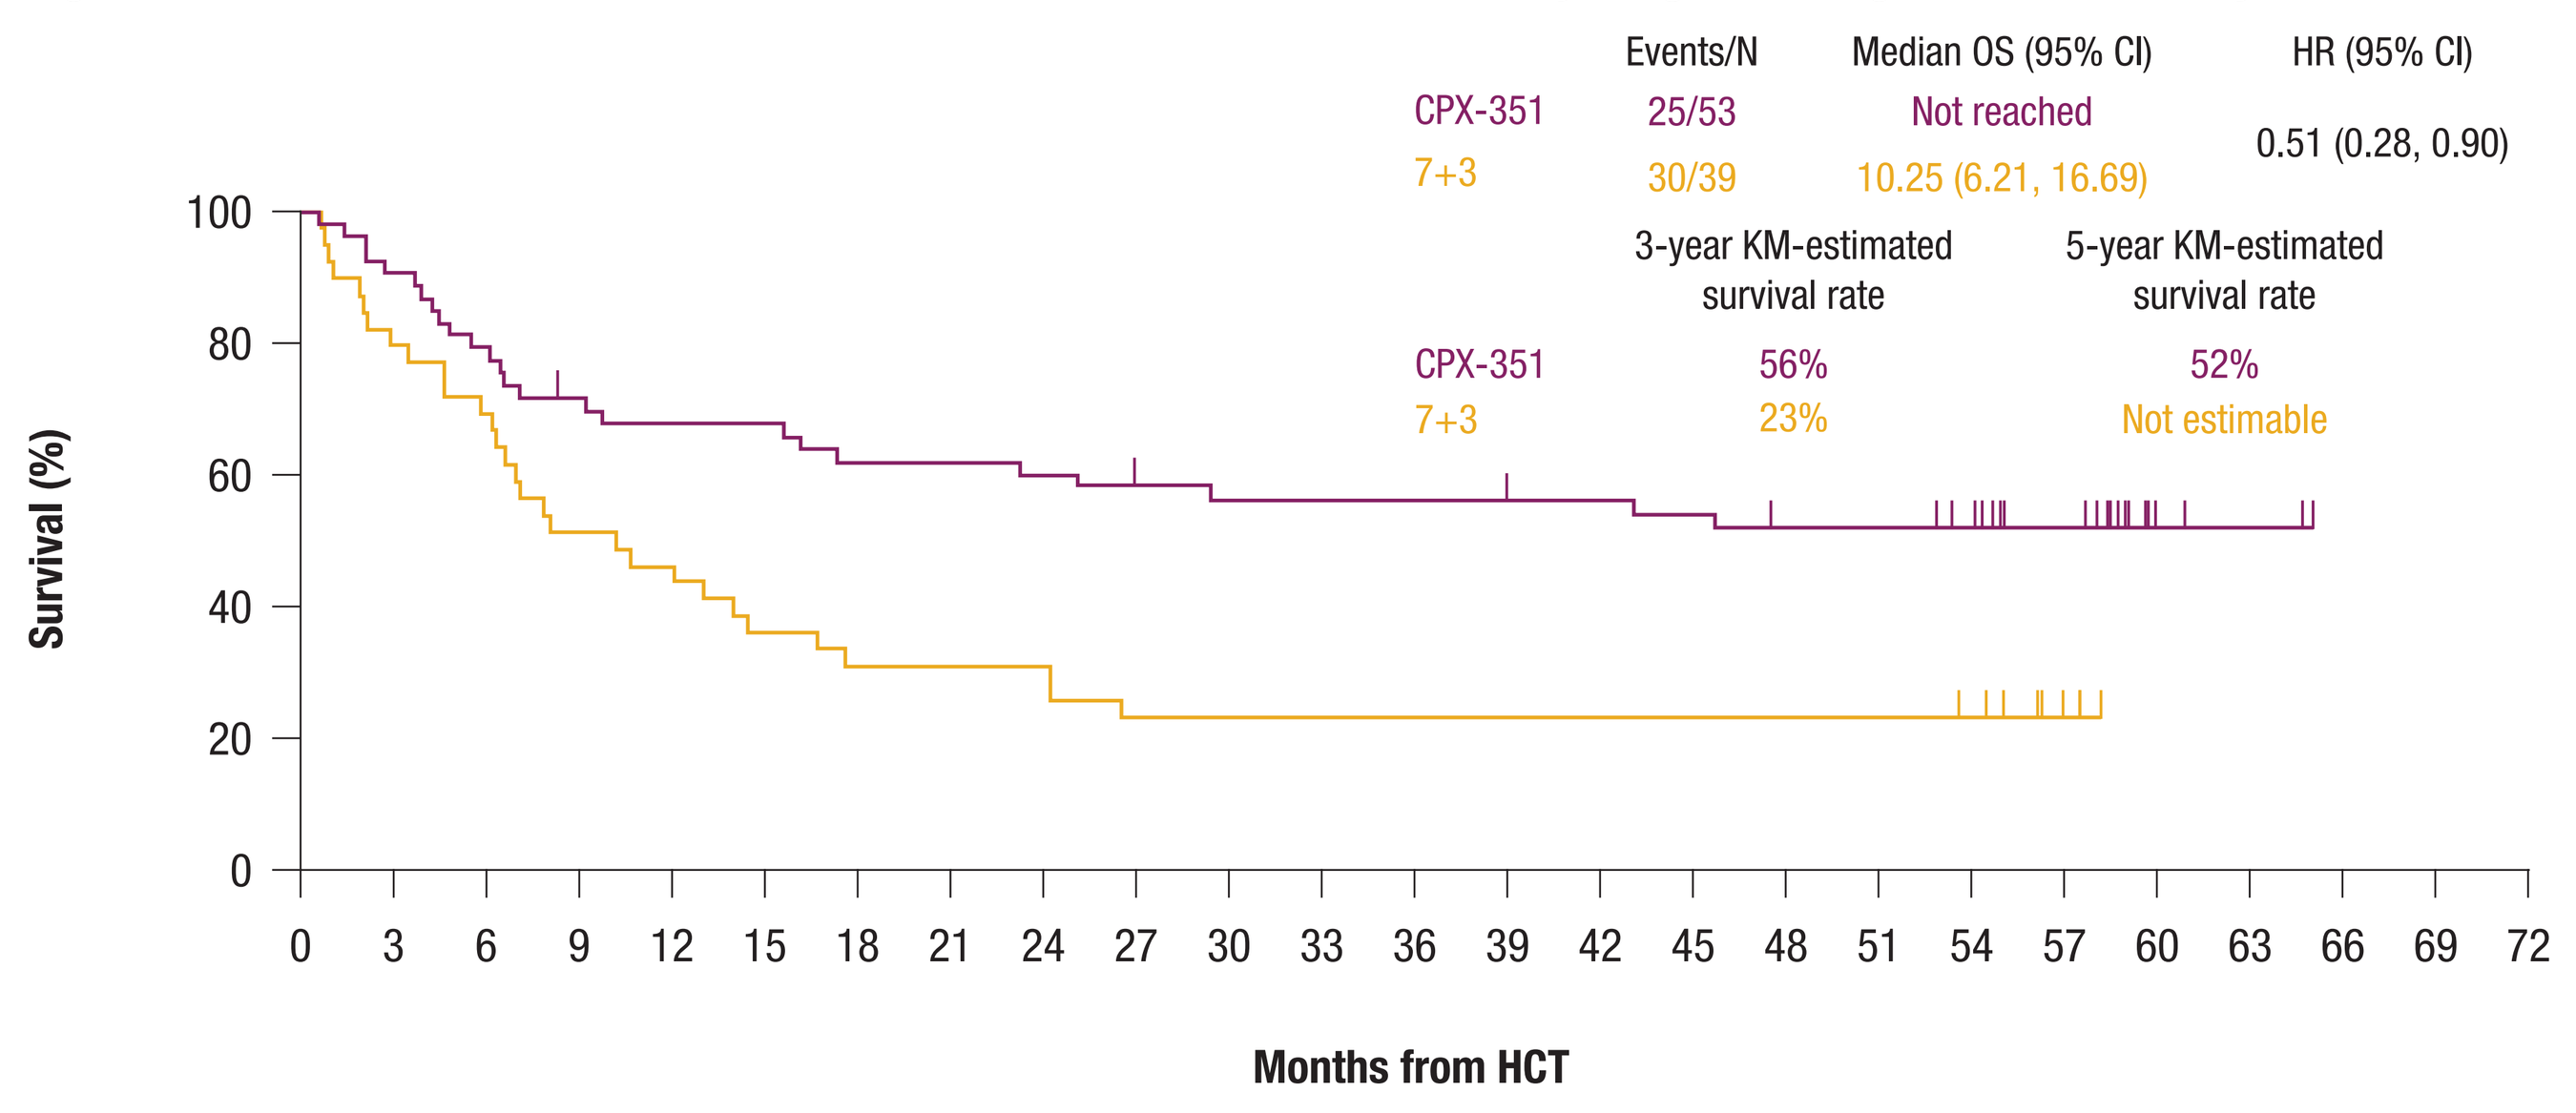

Events/N
Median OS (95% CI)
HR (95% CI)
CPX-351
25/53
Not reached
7+3
30/39
10.25 (6.21, 16.69)
0.51 (0.28, 0.90)
3-year KM-estimated survival rate
5-year KM-estimated survival rate
CPX-351
56%
52%
7+3
23%
Not estimable
Survival (%)
100
80
60
40
20
0
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51 54 57 60 63 66 69 72
Months from HCT

CPX-351	53	48	42	37	35	35	32	32	31	29	28	28	28	27	27	26	24	24	21	15	6	2	0	0	0
7+3	39	31	27	20	18	14	12	12	12	9	9	9	9	9	9	9	9	9	8	2	0	0	0	0	0

OS, overall survival; HCT, hematopoietic cell transplantation; AML, acute myeloid leukemia; CI, confidence interval; HR, hazard ratio; KM, Kaplan-Meier.

- HCT was received by 53/153 (35%) and 39/156 (25%) patients in the CPX-351 and 7+3 arms, respectively
- Median OS landmarked from the date of HCT was not reached for CPX-351 versus 10.25 months for 7+3 and the Kaplan-Meier–estimated survival rates landmarked from the date of HCT were higher for CPX-351 versus 7+3 at 3 and 5 years, and was >50% at 5 years for patients treated with CPX-351
- The majority of patients who underwent HCT were in CR or CRi in both the CPX-351 arm (41/53 [77%]) and the 7+3 arm (24/39 [62%])

Figure 3. OS in Older Adults With Newly Diagnosed High-risk/Secondary AML Who Achieved CR or CRi

A. All Patients Who Achieved CR or CRi

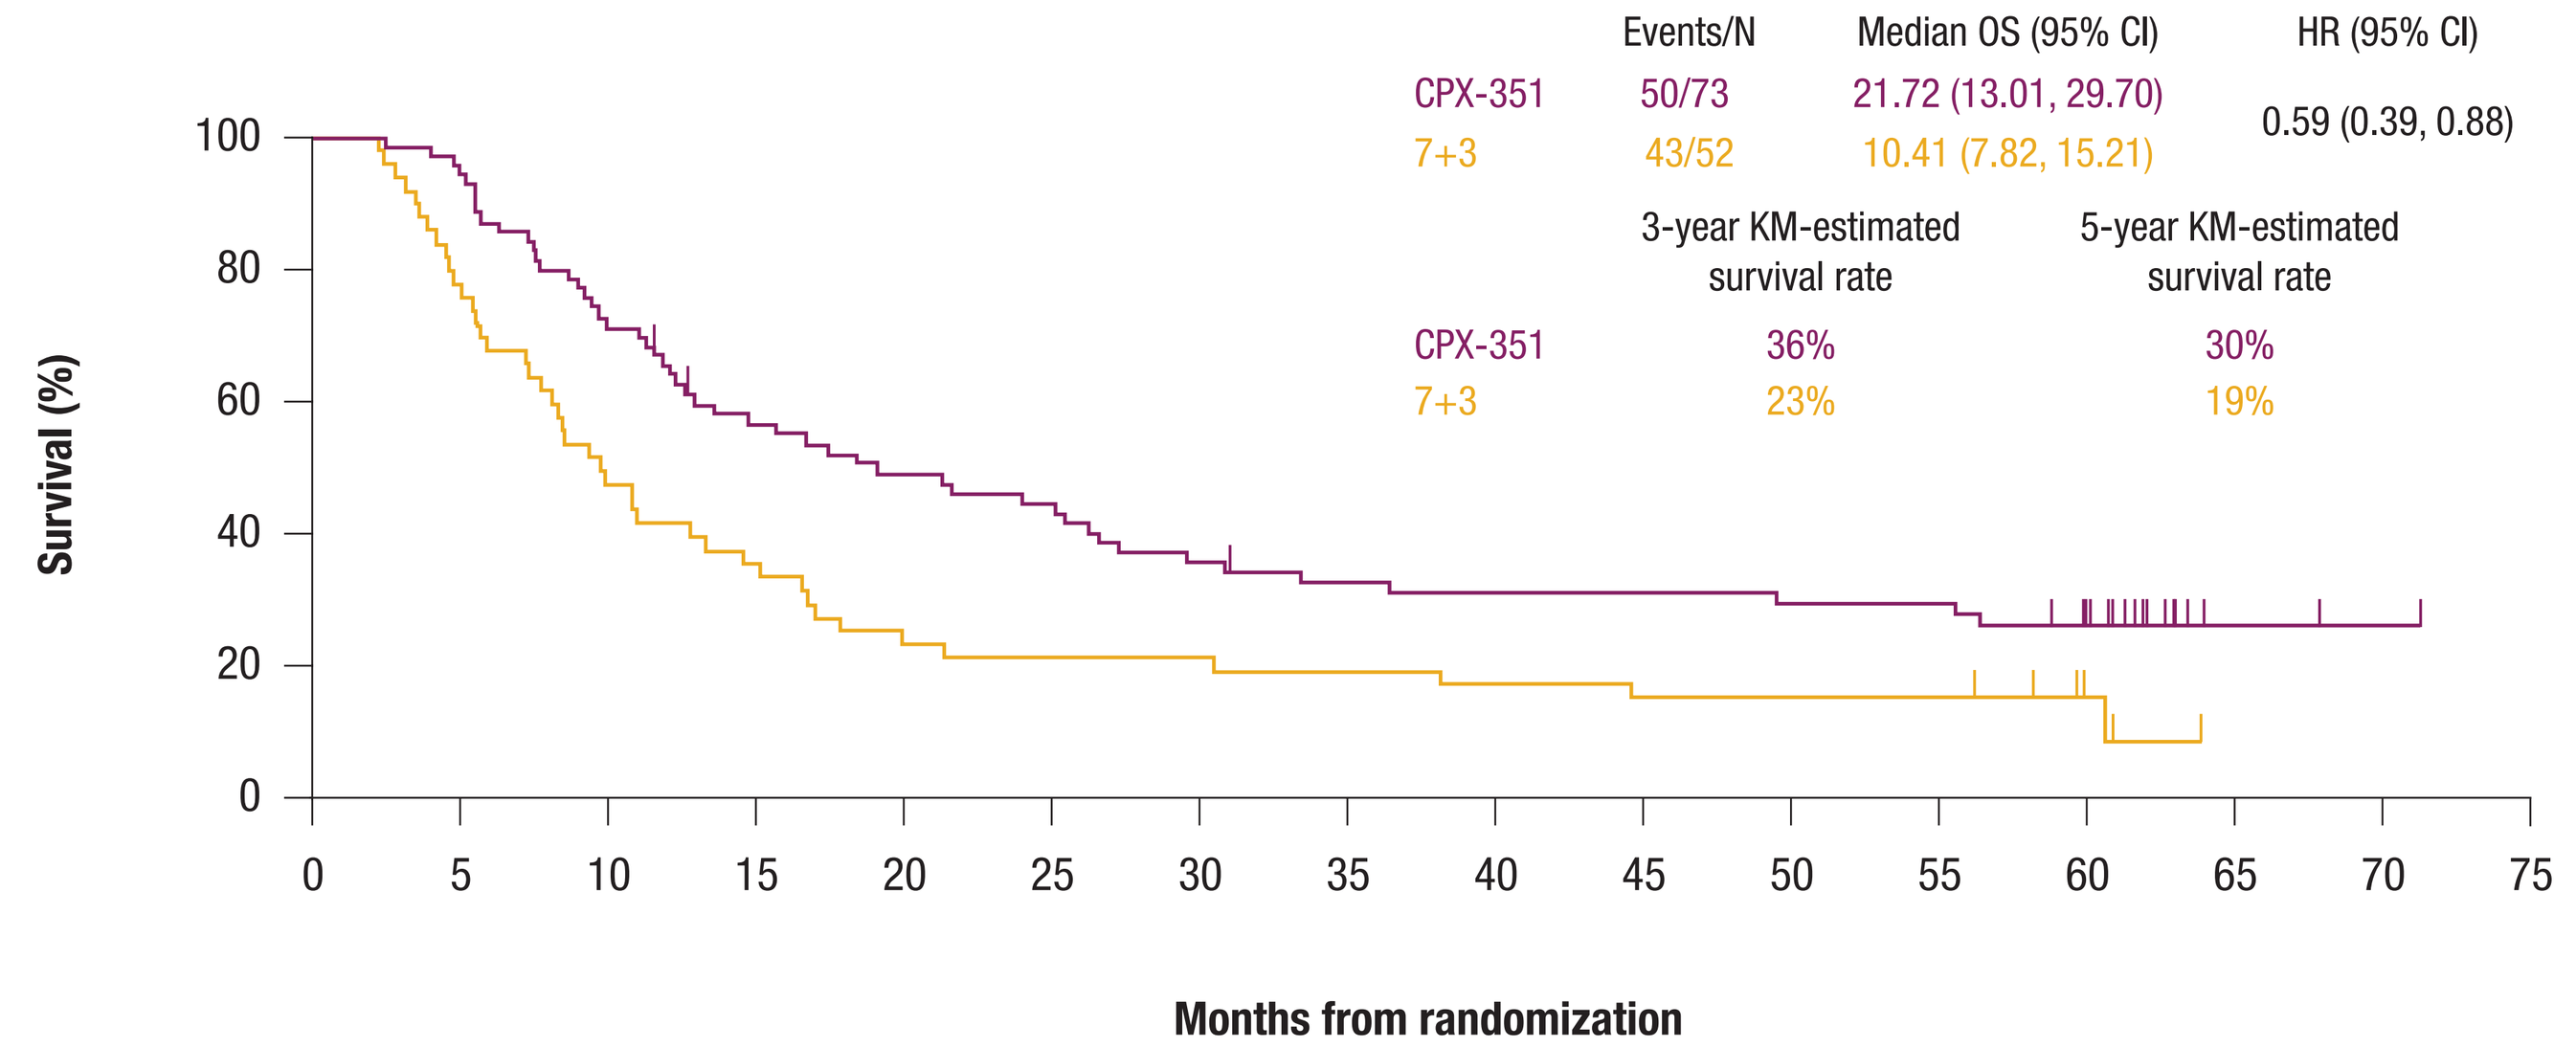

Events/N
Median OS (95% CI)
HR (95% CI)
CPX-351
50/73
21.72 (13.01, 29.70)
7+3
43/52
10.41 (7.82, 15.21)
0.59 (0.39, 0.88)
3-year KM-estimated survival rate
5-year KM-estimated survival rate
CPX-351
36%
30%
7+3
23%
19%
Survival (%)
100
80
60
40
20
0
0 5 10 15 20 25 30 35 40 45 50 55 60 65 70 75
Months from randomization

CPX-351	73	69	54	41	36	33	27	24	23	23	22	22	18	2	1	0
7+3	52	41	26	20	15	13	13	12	11	10	10	10	3	0	0	0

B. Patients Who Achieved CR or CRi and Underwent HCT (Landmarked From the Date of HCT)

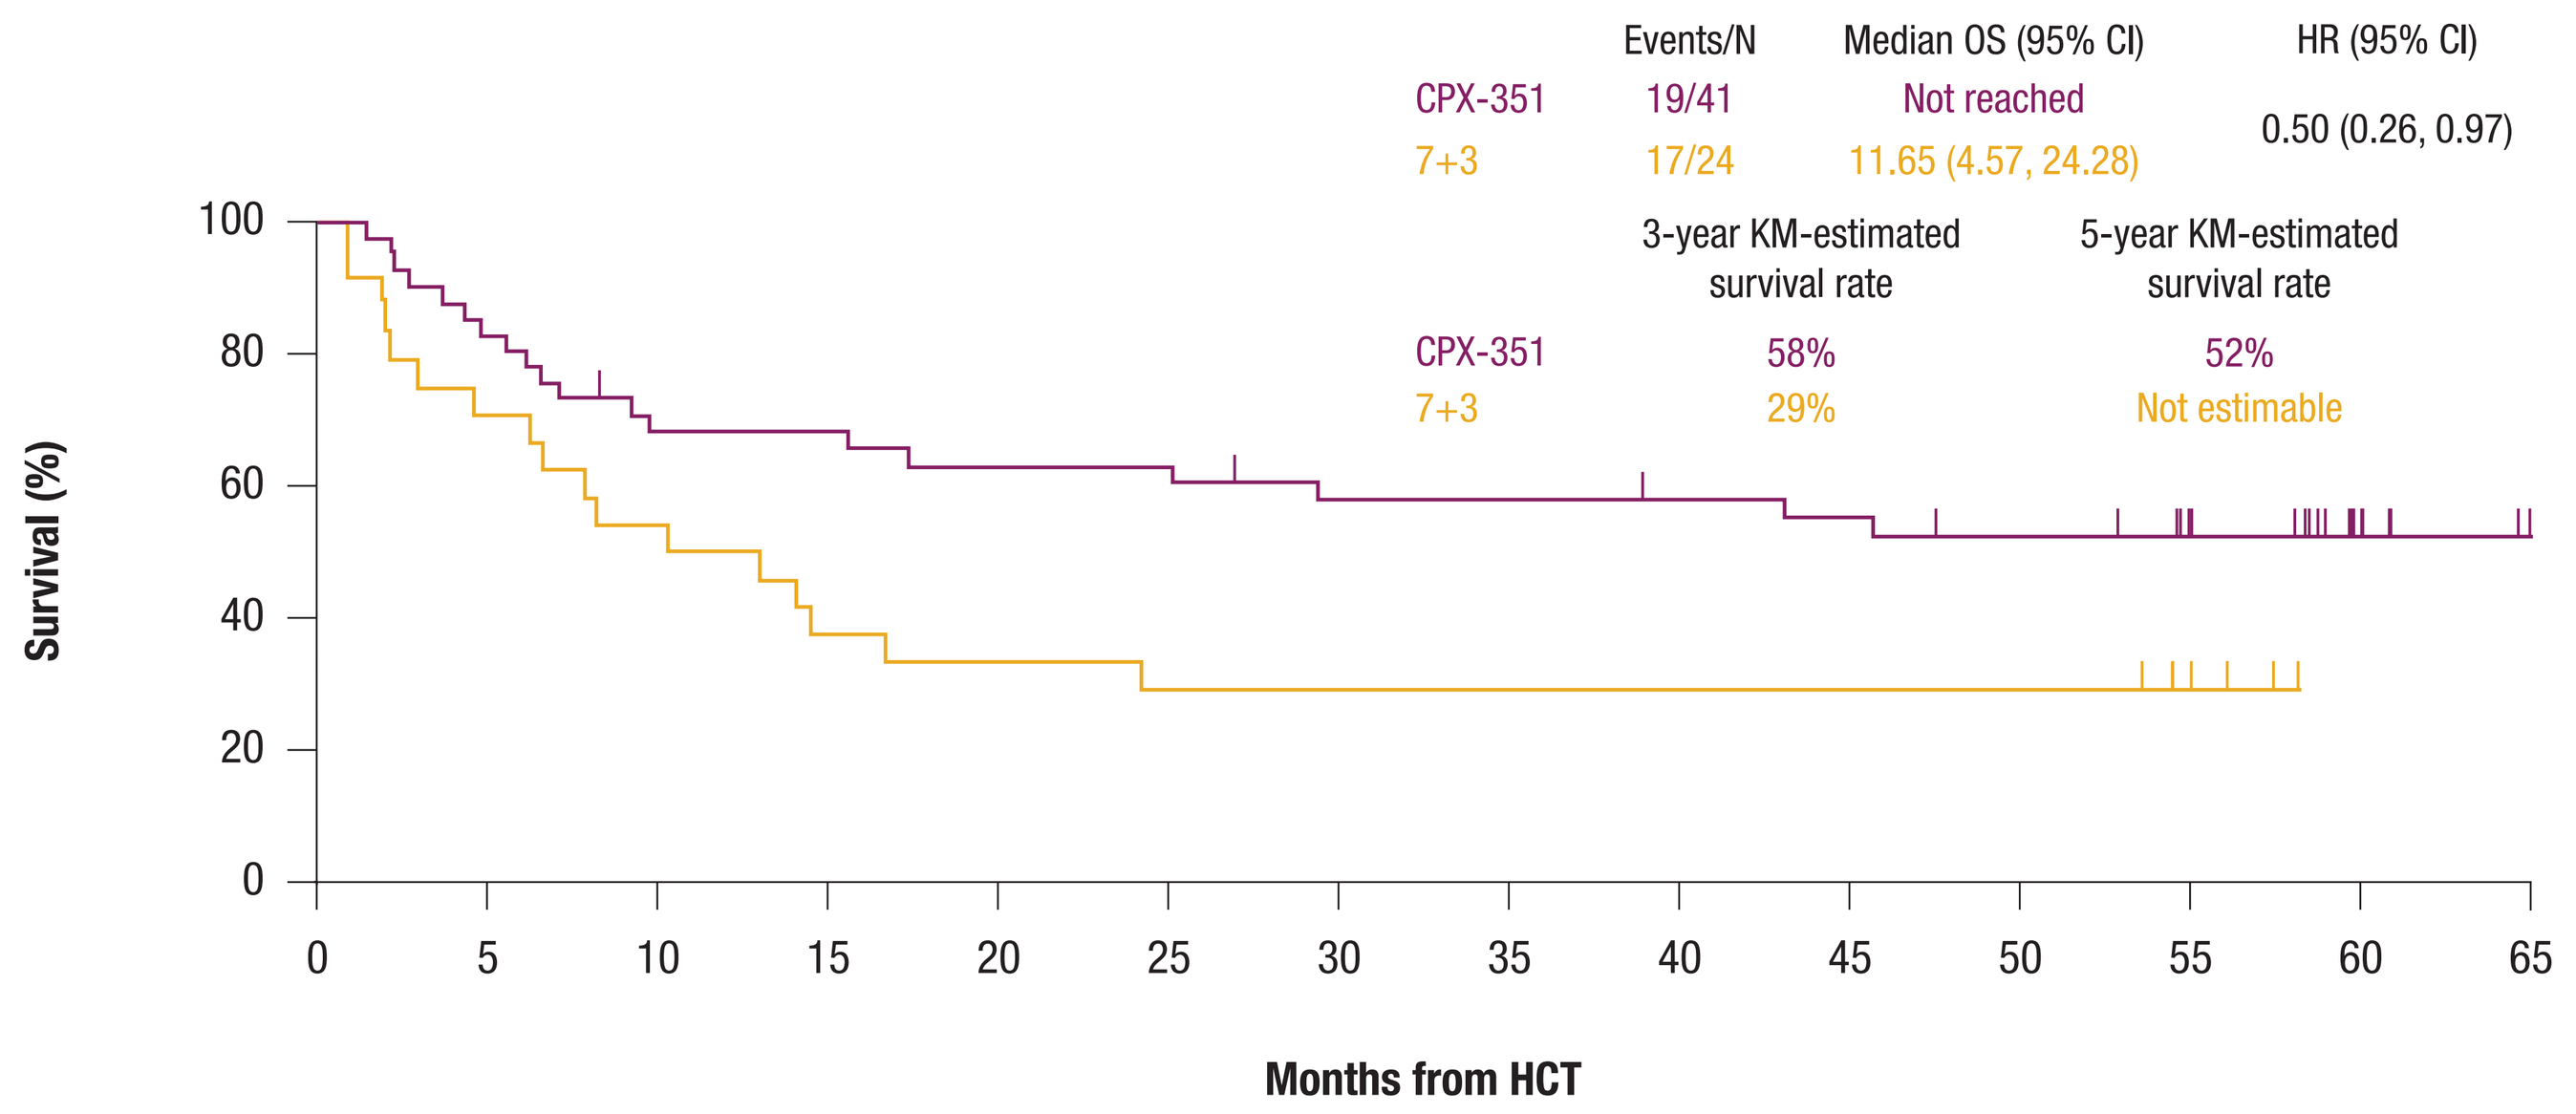

Events/N
Median OS (95% CI)
HR (95% CI)
CPX-351
19/41
Not reached
7+3
17/24
11.65 (4.57, 24.28)
0.50 (0.26, 0.97)
3-year KM-estimated survival rate
5-year KM-estimated survival rate
CPX-351
58%
52%
7+3
29%
Not estimable
Survival (%)
100
80
60
40
20
0
0 5 10 15 20 25 30 35 40 45 50 55 60 65
Months from HCT

CPX-351	41	34	27	27	25	25	22	22	21	20	18	13	6	0
7+3	24	17	13	9	8	7	7	7	7	7	7	5	0	0

OS, overall survival; AML, acute myeloid leukemia; CR, complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; CI, confidence interval; KM, Kaplan-Meier; HCT, hematopoietic cell transplantation.

- CR or CRi was achieved by 73 (48%) patients in the CPX-351 arm and 52 (33%) patients in the 7+3 arm
- CPX-351 consolidation was administered to 49 patients, including 1 patient who had not achieved a CR or CRi during induction, and 5+2 consolidation was administered to 32 patients who had achieved remission
 - Two consolidation cycles were received by 23 patients treated with CPX-351 versus 12 patients treated with 5+2
- Among all patients who achieved CR or CRi, median OS was longer with CPX-351 versus 7+3 and the Kaplan-Meier–estimated survival rate was higher for CPX-351 versus 7+3 at 3 and 5 years
- Among patients who achieved CR or CRi, 41/73 (56%) in the CPX-351 arm and 24/52 (46%) in the 7+3 arm subsequently underwent HCT

Table 3. Primary Cause of Death in Patients With Newly Diagnosed High-risk/Secondary AML

Deaths, n (%)	CPX-351 (n = 153)	7+3 (n = 151)
Total number of patients who died	124 (81)	140 (93)
Primary cause of death		
Progressive leukemia	70 (56)	74 (53)
Nonprogressive disease, cancer-related organ failure	0	5 (4)
Adverse event	17 (14)	19 (14)
Other	20 (16)	24 (17)
Unknown	17 (14)	18 (13)

AML, acute myeloid leukemia.

- Among the patients who died, causes of death were similar between arms, and the most common primary cause of death was progressive leukemia in both arms
- Early mortality rates for CPX-351 versus 7+3, respectively, were 6% versus 11% by Day 30 and 14% versus 21% by Day 60

Conclusions

- Final 5-year follow-up results from this phase 3 study in older adults with newly diagnosed high-risk/secondary AML demonstrated that improved OS with CPX-351 versus conventional 7+3 chemotherapy was maintained in the overall study population, as well as among patients who achieved CR or CRi and those who underwent HCT
- The longer OS with CPX-351 versus 7+3 in patients who achieved CR or CRi and in those who underwent HCT suggests potentially deeper responses may be achieved with CPX-351 treatment
- All-cause mortality and early mortality (30- and 60-day) rates were lower with CPX-351 versus 7+3
- These data support the prior evidence that CPX-351 has the ability to produce or contribute to long-term remission and survival in older patients with newly diagnosed high-risk/secondary AML

References: **1.** Granfeldt Østgård LS, et al. *J Clin Oncol.* 2015;33(31):3641-3649. **2.** Appelbaum FR, et al. *Blood.* 2006;107(9):3481-3485. **3.** Kayser S, et al. *Blood.* 2011;117(7):2137-2145. **4.** VYXEOS® (daunorubicin and cytarabine) liposome for injection, for intravenous use [package insert]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **5.** European Medicines Agency. Vyxeos liposomal 44 mg/100 mg powder for concentrate for solution for infusion. https://www.ema.europa.eu/en/medicines/human/EPAR/vyxeos-liposomal. Accessed April 7, 2020. **6.** Lancet JE, et al. *J Clin Oncol.* 2018;36(26):2684-2692.

Support: This study was supported by Jazz Pharmaceuticals.

Acknowledgments: Medical writing and editorial assistance were provided by Senem K. Lubin, PhD, of SciFluent Communications, Inc., and were financially supported by Jazz Pharmaceuticals.

Disclosures: **JEL** served as a consultant for Agios, Daiichi Sankyo, Jazz Pharmaceuticals, and Pfizer. **GLU** served as a consultant for Jazz Pharmaceuticals and Pfizer; and received honoraria from Astellas Pharma. **EKR** served as a consultant for Celgene, Incyte, Novartis, and Pfizer; served on speakers bureaus for Ariad, Celgene, Incyte, and Novartis; received research funding from Astellas Pharma, Bristol-Myers Squibb, Novartis, NS Pharma, and Pfizer; and received travel support from Celgene and Novartis. **RKS** served as a consultant for Sunesis; received research funding from Agios, Astellas Pharma, Bayer, Incyte, Jazz Pharmaceuticals, and Sunesis; received honoraria from Sunesis; and received travel support from Sunesis. **SAS** served as a consultant for Astellas Pharma, Baxalta, Boehringer Ingelheim, CTI BioPharma, Jazz Pharmaceuticals, Kite Pharma, Sunesis, and Tolero; and received institutional research funding from Boehringer Ingelheim and Sunesis. **DLB** received research funding from GlycoMimetics. **JEK** received honoraria from Gilead, Magellan, and Novartis; consulting fees from Gilead, Magellan, Novartis, Pharmacyclics, and Seattle Genetics; research funding from Boehringer Ingelheim, Cantex, Erytech, and Millennium; and travel support from Gilead, Novartis, and Seattle Genetics. **GJS** received research funding from AbbVie, Agios, Actinium, Amgen, Ariad, Astellas Pharma, Bristol-Myers Squibb, Celator/Jazz Pharmaceuticals, Constellation, Cyclacel, Daichii Sankyo, Deciphera, Delta-Fly, Forma, FujiFilm, Gamida, Genentech-Roche, Geron, Incyte, Karyopharm, Kite Pharma, Mateon, MedImmune, Novartis, Onconova, Pfizer, Regimmune, Samus, Sangamo, Tolero, and Trovagene; served as a consultant for Agios, Amgen, AstraZeneca, Incyte, Novartis, and Ono Pharma; served on speakers bureaus for Agios, Amgen, Celgene, Gilead, Incyte, Sanofi, and Stemline; provided expert testimony for Kaiser Permanente; and holds stock in Amgen, Bristol-Myers Squibb, Johnson & Johnson, and Pfizer. **MJW** received research funding from Amgen, Leadiant, Merck, and Shire; participated in advisory committees for Daiichi Sankyo; and holds stock ownership in Reata Pharmaceuticals. **DHR** holds stock ownership in AbbVie and patents/royalties with the University of Rochester. **SF** and **YLC** are employees of and hold stock in Jazz Pharmaceuticals. **JEC** served as a consultant for Astellas Pharma, Daiichi Sankyo, Jazz Pharmaceuticals, Novartis, and Pfizer; and received research funding from Arog, Astellas Pharma, Daiichi Sankyo, Jazz Pharmaceuticals, Novartis, and Pfizer. **LFN**, **TLL**, **DH**, and **SRS** have no relationships to disclose.

Contact information: For questions or comments, please contact Jeffrey E. Lancet at Jeffrey.Lancet@moffitt.org.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.